Potent Antioxidant and Mitochondrial-protectant Effects of ATH434, a Novel Inhibitor of α-Synuclein Aggregation with Moderate Iron-binding Affinity

Danielle K. Bailey[1], Rhudwan Nihlawi[1], Margaret J. Bradbury[2], Daniel J. Kosman[1]

[1] Department of Biochemistry, SUNY University at Buffalo, Buffalo, NY

[2] Alterity Therapeutics, Newark, CA

Introduction

Iron is essential for supporting energy metabolism, mitochondrial function, and maintaining cellular redox potential. Excess labile iron can generate reactive oxygen species in mitochondria which, if unchecked, can lead to sustained oxidative stress and eventual cell death. Parkinson's disease (PD) and Multiple System Atrophy (MSA) are neurodegenerative conditions characterized by regional excess brain iron and resultant oxidative stress in areas of pathology, leading to clinical trials of iron binding small molecules for their treatment. ATH434, a small molecule drug candidate with moderate ferric iron affinity (K_d 10^{-10}) [1], promotes cellular iron efflux, reduces excess brain iron and aggregated α-synuclein, improves neuronal survival, and restores motor performance in murine PD and MSA models. ATH434 is currently in phase 2 MSA trials. Deferiprone (DFP) is a high ferric iron affinity drug (K_d 10^{-21}) [2,3] approved for treating systemic iron-overload disorders. Because DFP is designed to reduce cellular iron stores, it has potential for maladaptive pharmacological effects in healthy cells [4]. DFP has also demonstrated efficacy in preclinical PD models. The required doses, however, are higher than expected given its ready brain access and high ferric iron affinity, suggesting that ATH434 may possess unique beneficial properties.

In this study, we investigated the efficacy of ATH434 and DFP as potential antioxidants and mitochondrial protectants using a menadione-induced model of oxidative stress in the glutamatergic neuronal HT22 cell line. We assessed both *in cellulo* and *in solution* superoxide and peroxide scavenging abilities of 434 in relation to it's non-iron binding analog, 434-met, as well as the known iron chelation therapeutics deferasirox, Dfx, and deferiprone, Dfp. We also determined the specific reducing potential by cyclic voltammetry, as well as electron transfer (ET) and hydrogen atom transfer (HAT) capacities of these compounds using the standard antioxidant assays FRAP and ORAC, respectively.

Methods

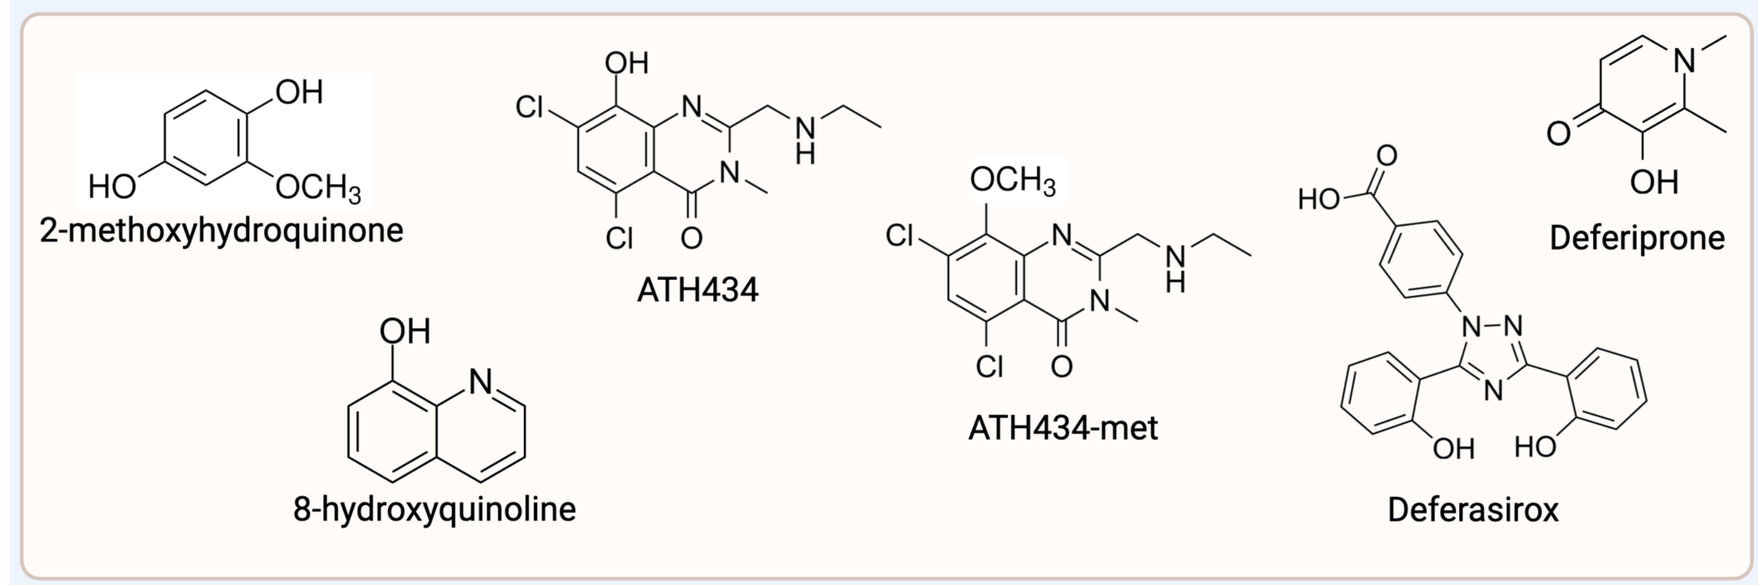
2-methoxyhydroquinone
ATH434
ATH434-met
Deferiprone
8-hydroxyquinoline
Deferasirox

Structures of chelator and antioxidant compounds used in this study.

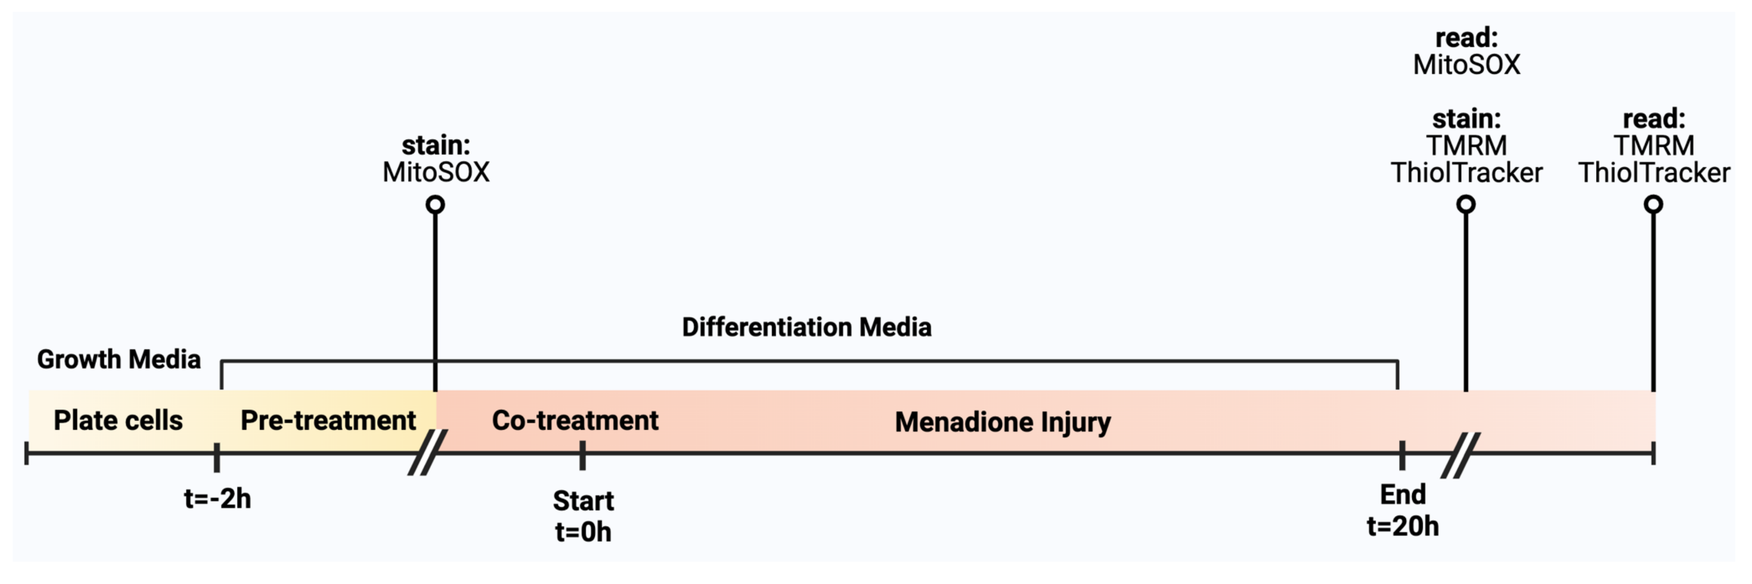
read: MitoSOX
stain: MitoSOX
stain: TMRM ThiolTracker
read: TMRM ThiolTracker
Growth Media
Differentiation Media
Plate cells
Pre-treatment
Co-treatment
Menadione Injury
t=-2h
Start t=0h
End t=20h

Neuronal menadione injury model timeline. *HT22 cells, an immortalized mouse hippocampal neuron line, were maintained in DMEM +10% FBS, 1X pen/strep, and 2mM L-Gln. Cells were plated on poly-D-lysine coated plates in growth media, then switched to Neurobasal + 1X N2 and 2mM L-Gln to differentiate for up to 24h. For fluorescence staining assays, cells were stained for 30min then washed 2X with HBSS prior to reading or continued treatments. Fluorescence was measured using BMG Labtech Fluostar Omega or BioTek Cytation 5.*

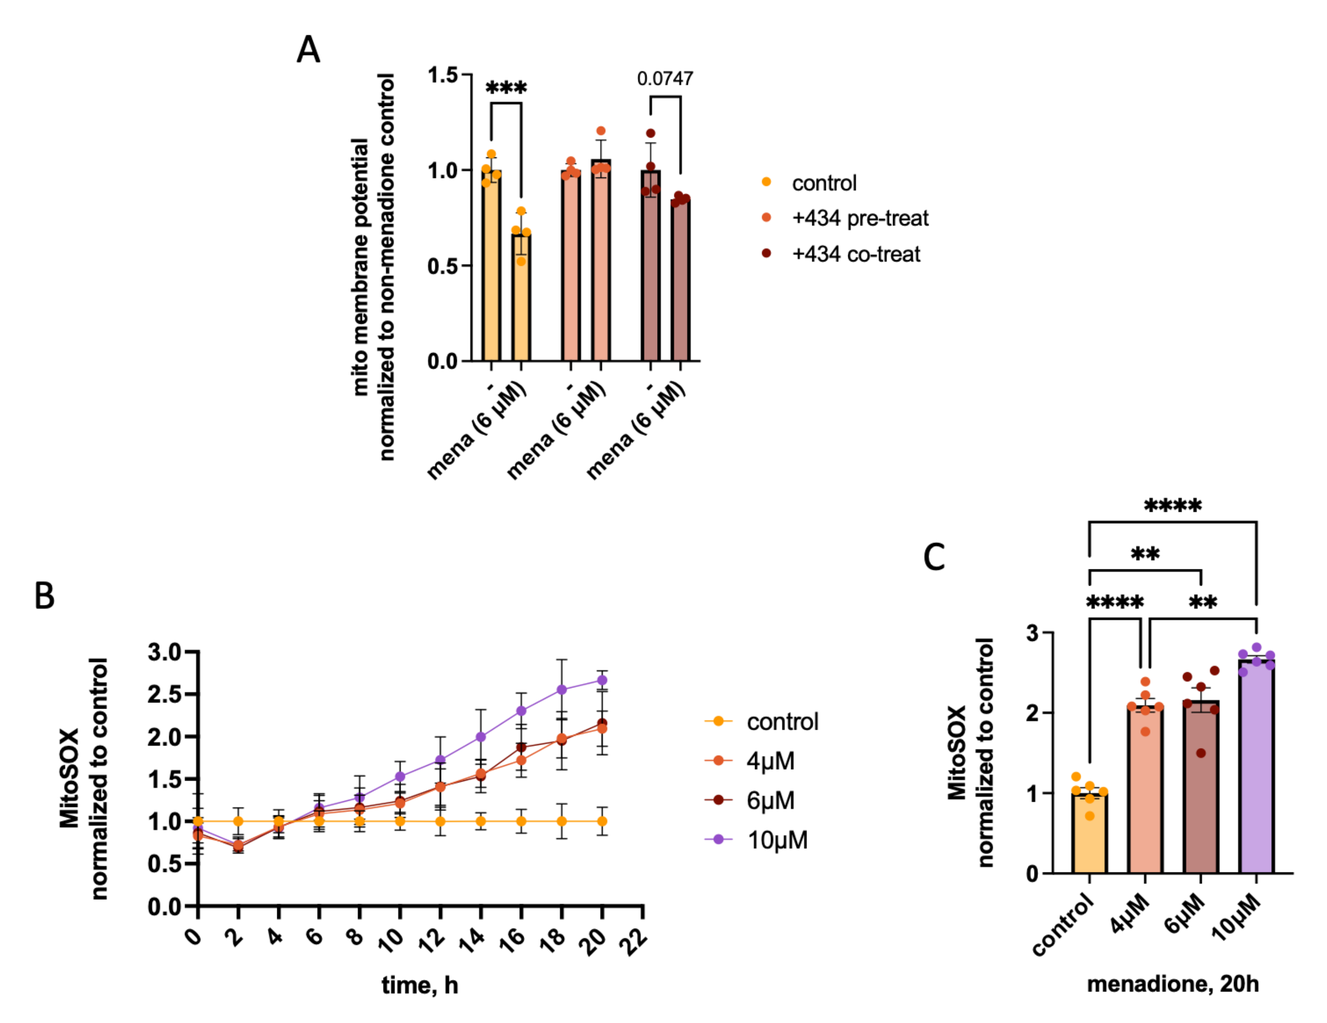
A
mito membrane potential normalized to non-menadione control
control
+434 pre-treat
+434 co-treat
B
MitoSOX normalized to control
time, h
control
4µM
6µM
10µM
C
menadione, 20h

Optimization of neuronal menadione injury model. A*, Mitochondrial membrane potential determined by TMRM assay using 6µM menadione for 20h with or without 20µM 434 co-treatment or pre-treatment for the first 2h.* ***B****, Optimization of timing of menadione injury in MitoSOX assay, up to 20h with increasing concentrations of menadione.* ***C****, Concentration dependence of menadione injury at 20h in MitoSOX assay.*

Results

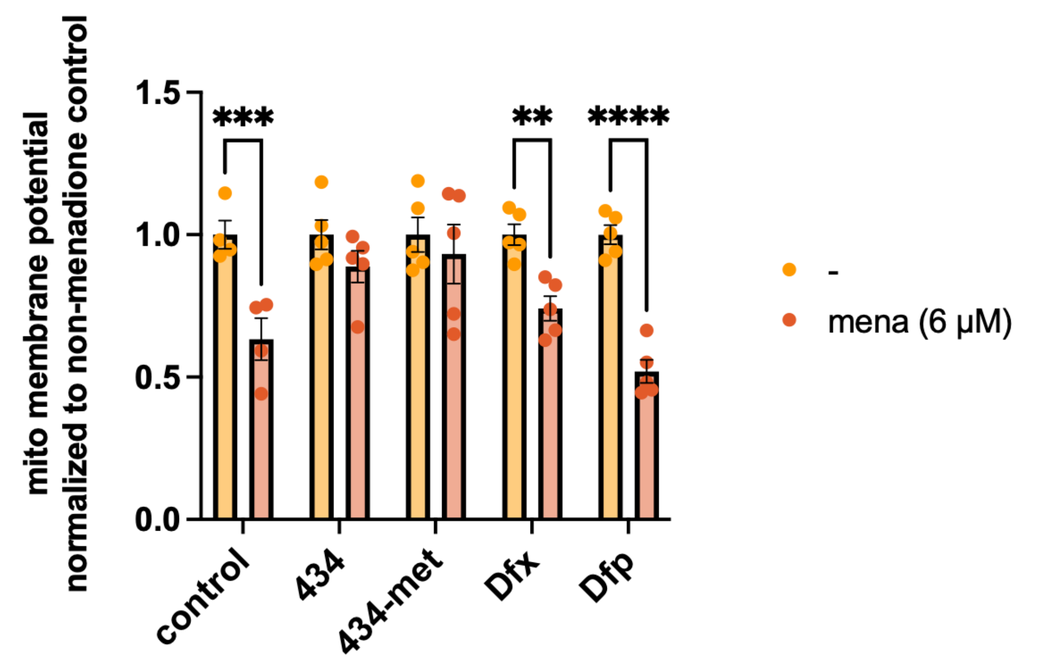
mito membrane potential normalized to non-menadione control
control
434
434-met
Dfx
Dfp
-
mena (6 µM)

434 and 434-met rescue menadione-induced loss of mitochondrial membrane potential. *TMRM assay was used to assess mitochondrial protective effects using 6µM menadione for 20h with or without 20µM compound pre-treatment for the first 2h. Data are expressed relative to MitoTracker Green and Hoechst, then normalized to non-menadione control in each treatment group.*

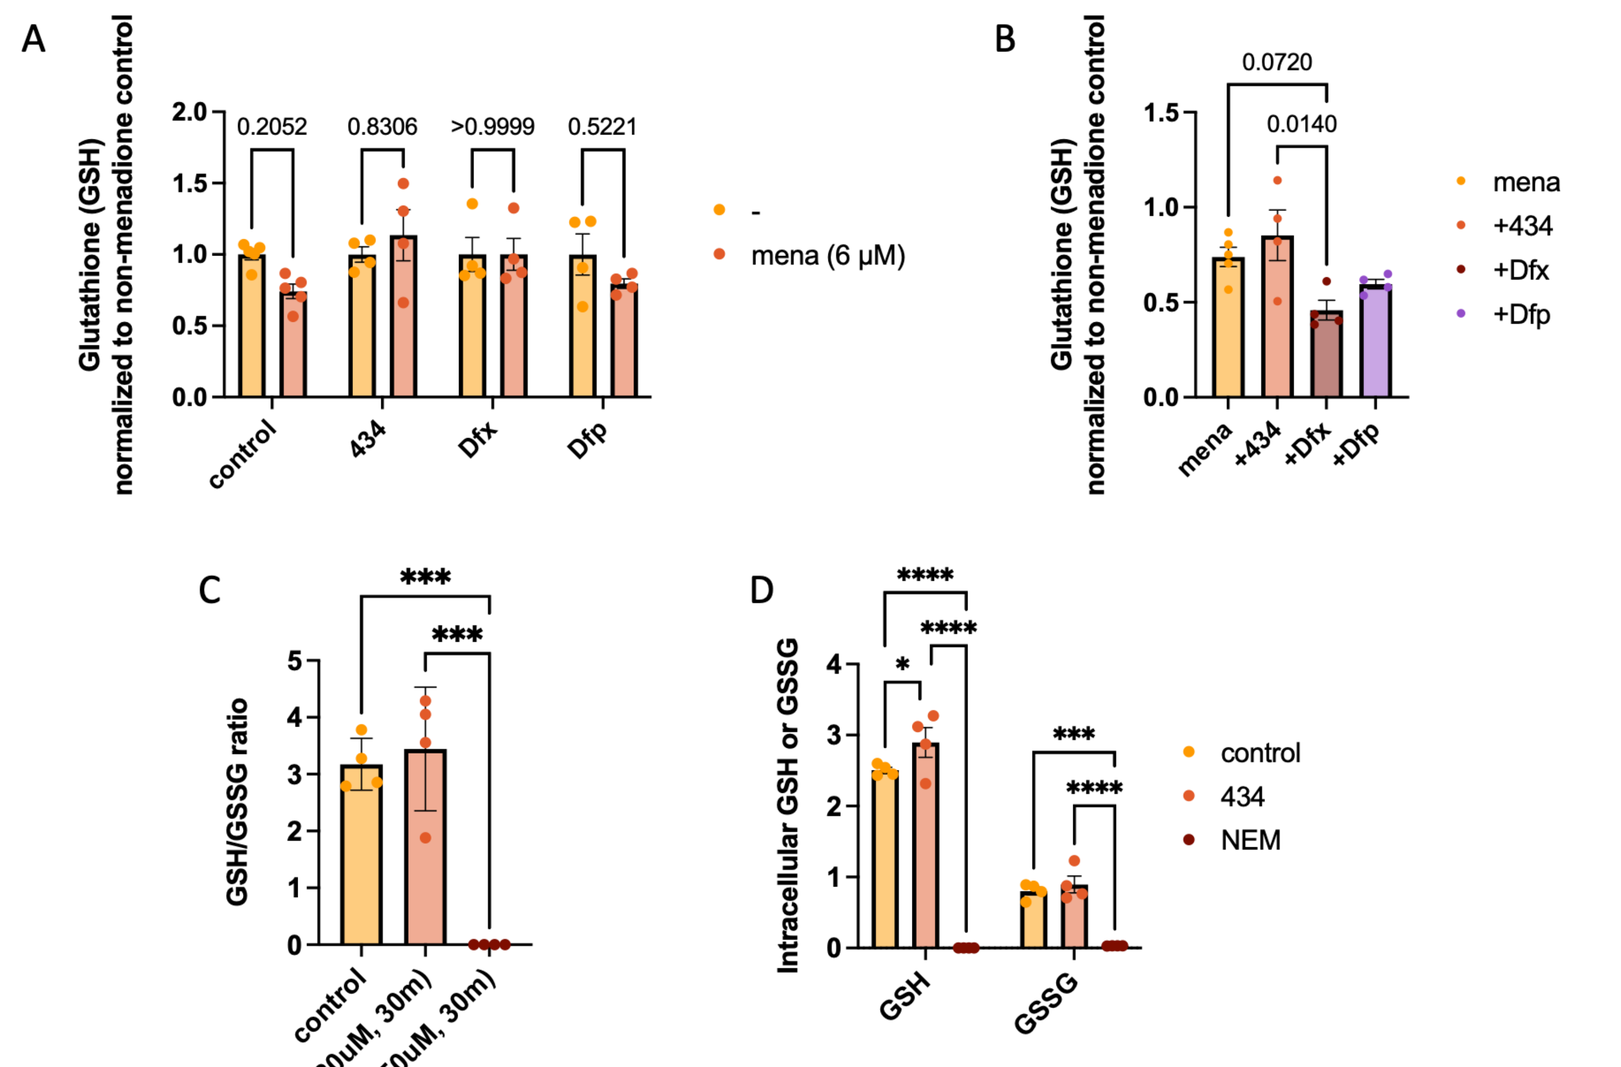
A
Glutathione (GSH) normalized to non-menadione control
B
C
GSH/GSSG ratio
D
Intracellular GSH or GSSG
control
434
NEM

Menadione-induced reduction in glutathione levels is not further altered by 434. A*, Intracellular GSH levels measured by ThiolTracker Violet, normalized to non-menadione control in each treatment group.* ***B****, GSH levels in menadione-treated cells, normalized to the untreated non-menadione control.* ***C****, Quantification GSH/GSSG ratio in cells treated with 434 or N-ethylmaleimide using GSH/GSSG ratio detection kit (abcam).* ***D****, Quantification of specifically reduced (GSH) and oxidized (GSSG) glutathione levels in cells. NEM depletes both GSH and GSSG levels in cells, while 434 does not.*

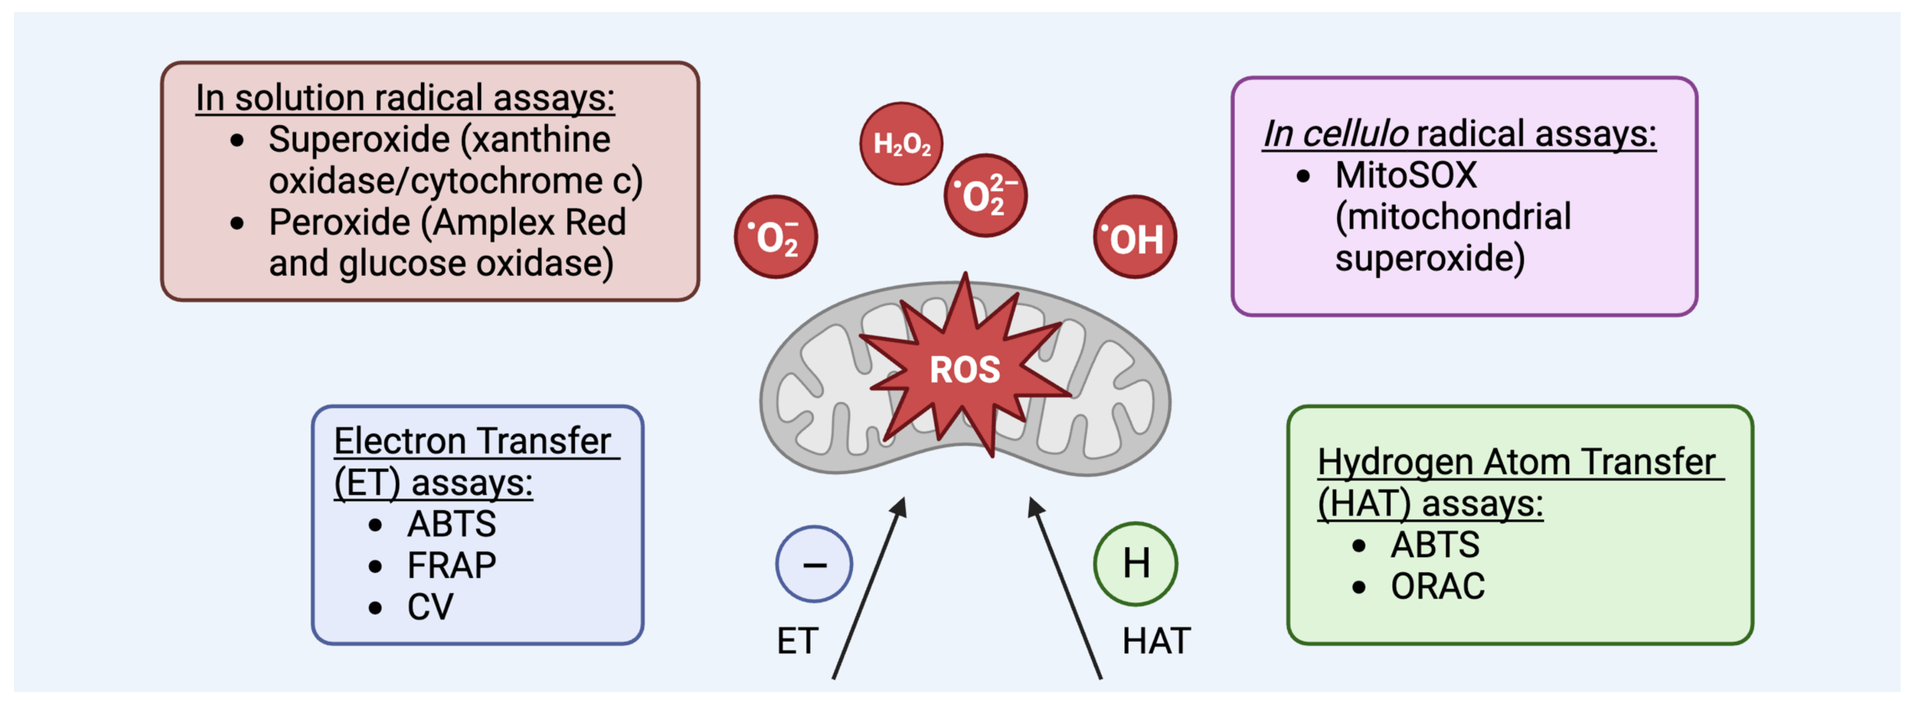
In solution radical assays:
Superoxide (xanthine oxidase/cytochrome c)
Peroxide (Amplex Red and glucose oxidase)
In cellulo radical assays:
MitoSOX (mitochondrial superoxide)
ROS
Electron Transfer (ET) assays:
ABTS
FRAP
CV
Hydrogen Atom Transfer (HAT) assays:
ABTS
ORAC
ET
HAT

Illustration of antioxidant assays used in this study. *Potential antioxidants were assessed for radical scavenging in both in solution and in cellulo assays, and antioxidant activity was assessed specifically for electron transfer (ET), hydrogen atom transfer (HAT), or both.*

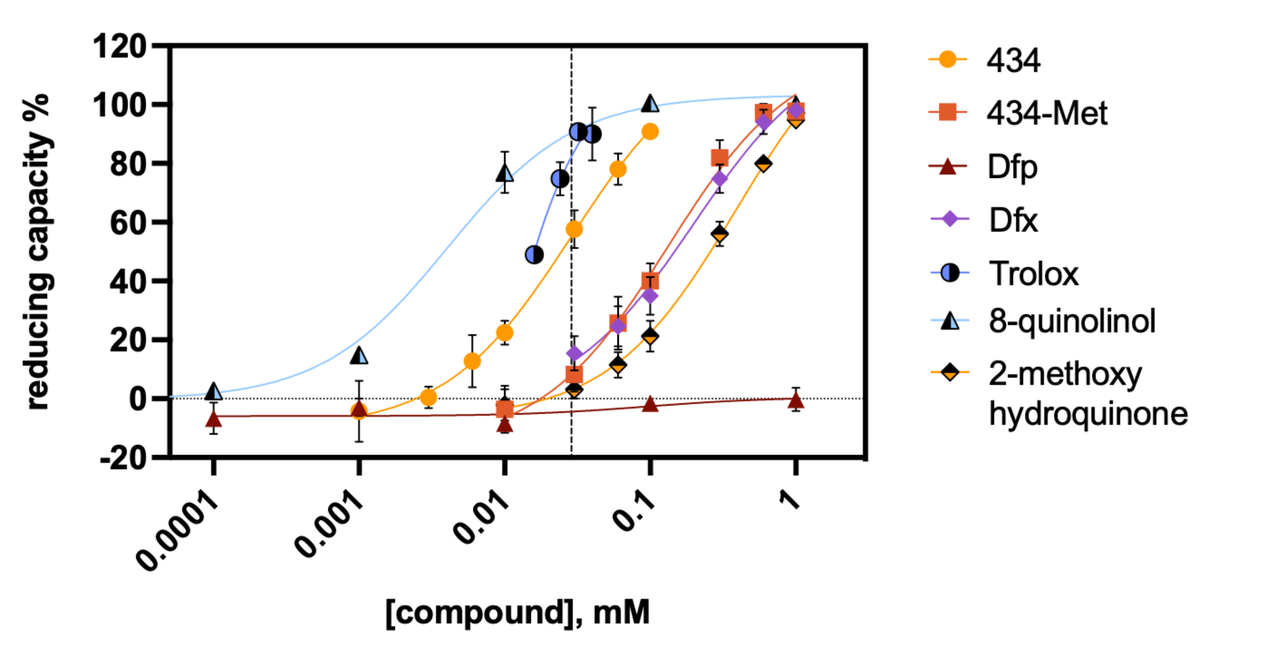
reducing capacity %
[compound], mM
434
434-Met
Dfp
Dfx
Trolox
8-quinolinol
2-methoxy hydroquinone

Compound	EC_{50} (µM) ± SEM
434	28.67 ± 4.12
434-met	16.04 ± 12.11
Dfp	Not calculable
Dfx	19.79 ± 35.83
Trolox	0.01 ± 7.65
2-methoxyhydroquinone	387.3 ± 39.4
8-quinolinol	4.05 ± 0.44

ATH434 has antioxidant activity while Dfp does not. *Using the ABTS assay, the concentration-dependent reducing capacity of each compound was determined. Data were fit using non-linear regression and the EC_{50} was determined and noted in the table (right). In addition to the compounds used in the above assays, the antioxidant Trolox (Vitamin E analog) and compounds with known reducing potentials, 8-hydroxyquinoline and 2-methoxyhydroquinone, were used for comparison. ABTS assesses both electron transfer (ET) and hydrogen atom transfer (HAT) activity.*

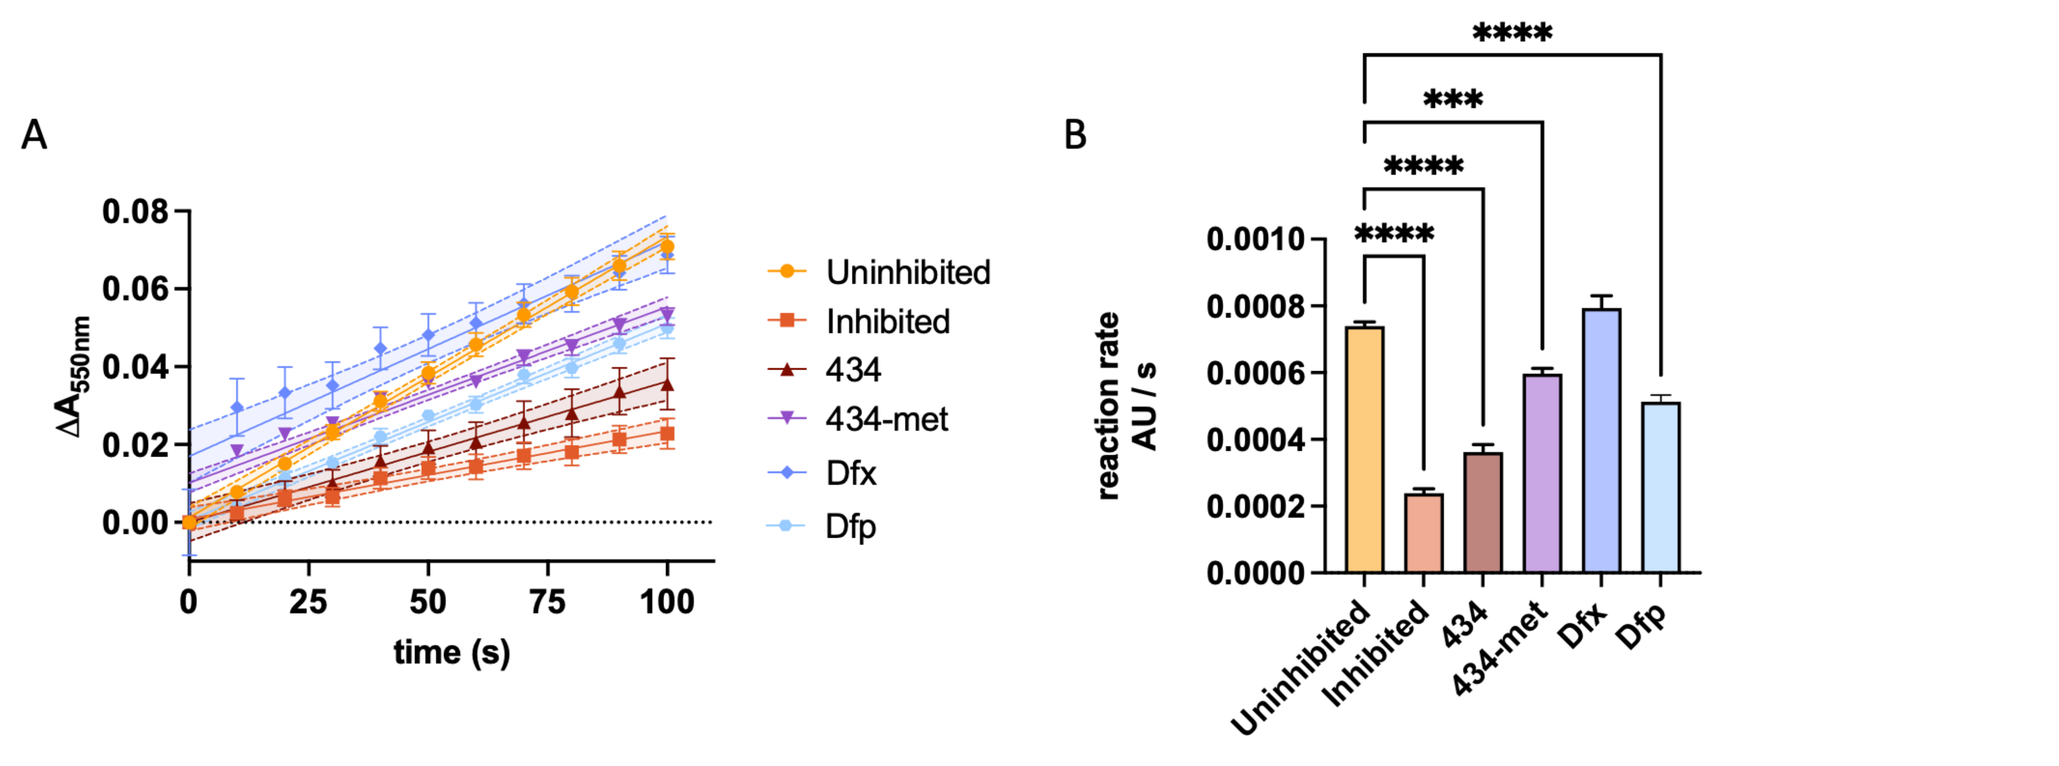
A
time (s)
Uninhibited
Inhibited
434
434-met
Dfx
Dfp
B
reaction rate AU / s

434 and 434-met inhibit superoxide production in solution. A*, Quantification of superoxide production over time in solution using Xanthine oxidase/cytochrome c detection. Inhibited, Superoxide dismutase (SOD) enzyme added to reaction. All compounds were used at 20µM final.* ***B****, Comparison of reaction rates for superoxide production, compared to uninhibited reaction.*

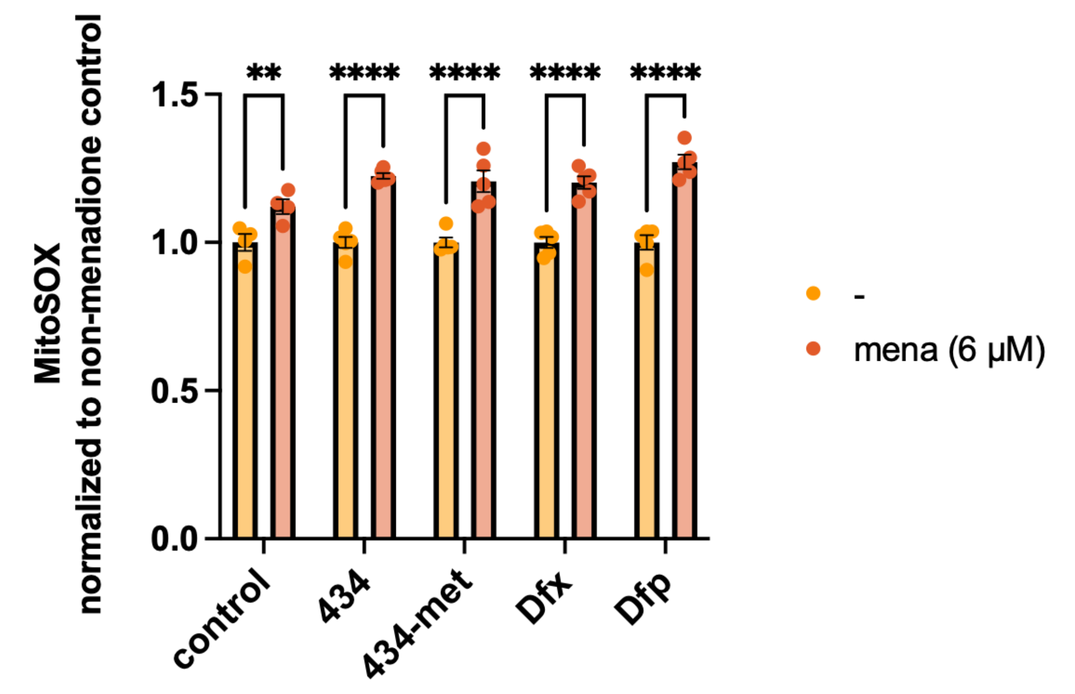
MitoSOX normalized to non-menadione control
-
mena (6 µM)

Menadione-induced superoxide accumulation in vivo was not rescued. *Mitochondrial superoxide accumulation was determined by MitoSOX assay using 6µM menadione for 20h with or without 20µM compound pre-treatment for the first 2h. Data are expressed relative to MitoTracker Green and Hoechst, then normalized to non-menadione control in each treatment group.*

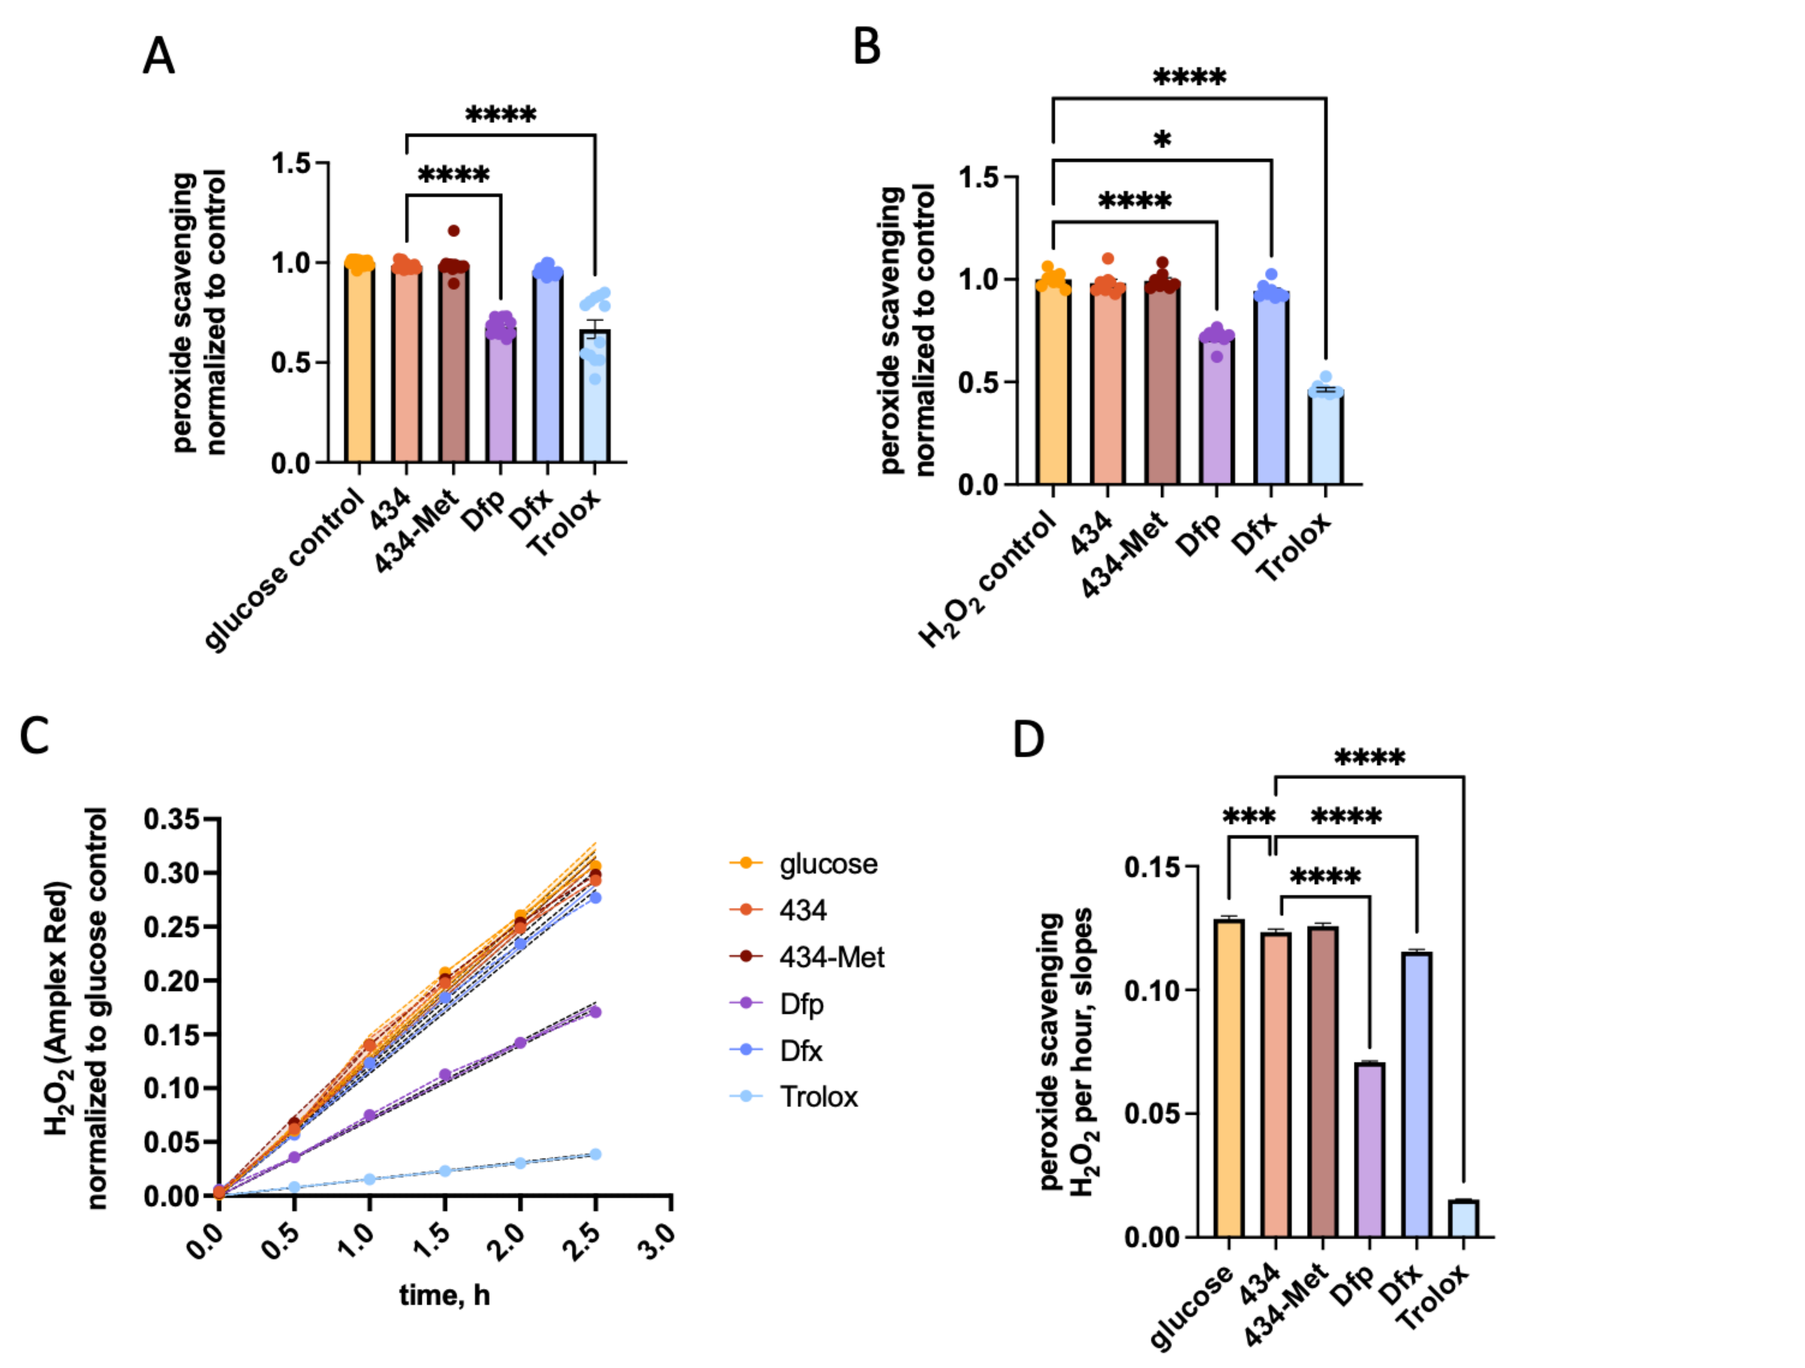
A
peroxide scavenging normalized to control
B
C
time, h
glucose
434
434-Met
Dfp
Dfx
Trolox
D
peroxide scavenging per hour, slopes

ATH434 does not directly scavenge peroxide. *Amplex Red was used as a reporter for H_2O_2 remaining in solution.* ***A****, Using glucose oxidase to continually generate peroxide, the peroxide scavenging of each compound at 20µM was assessed.* ***B****, To confirm whether the Dfp results were due to peroxide scavenging and not direct enzyme inhibition, compounds were incubated with 100µM H_2O_2 for 30min prior to assay, then incubated with Amplex Red.* ***C****, Quantification of peroxide scavenging over time. Compounds were incubated as in panel A and absorbance was measured every 30min for 2.5h.* ***D****, Comparison of peroxide scavenging rates, compared to 434.*

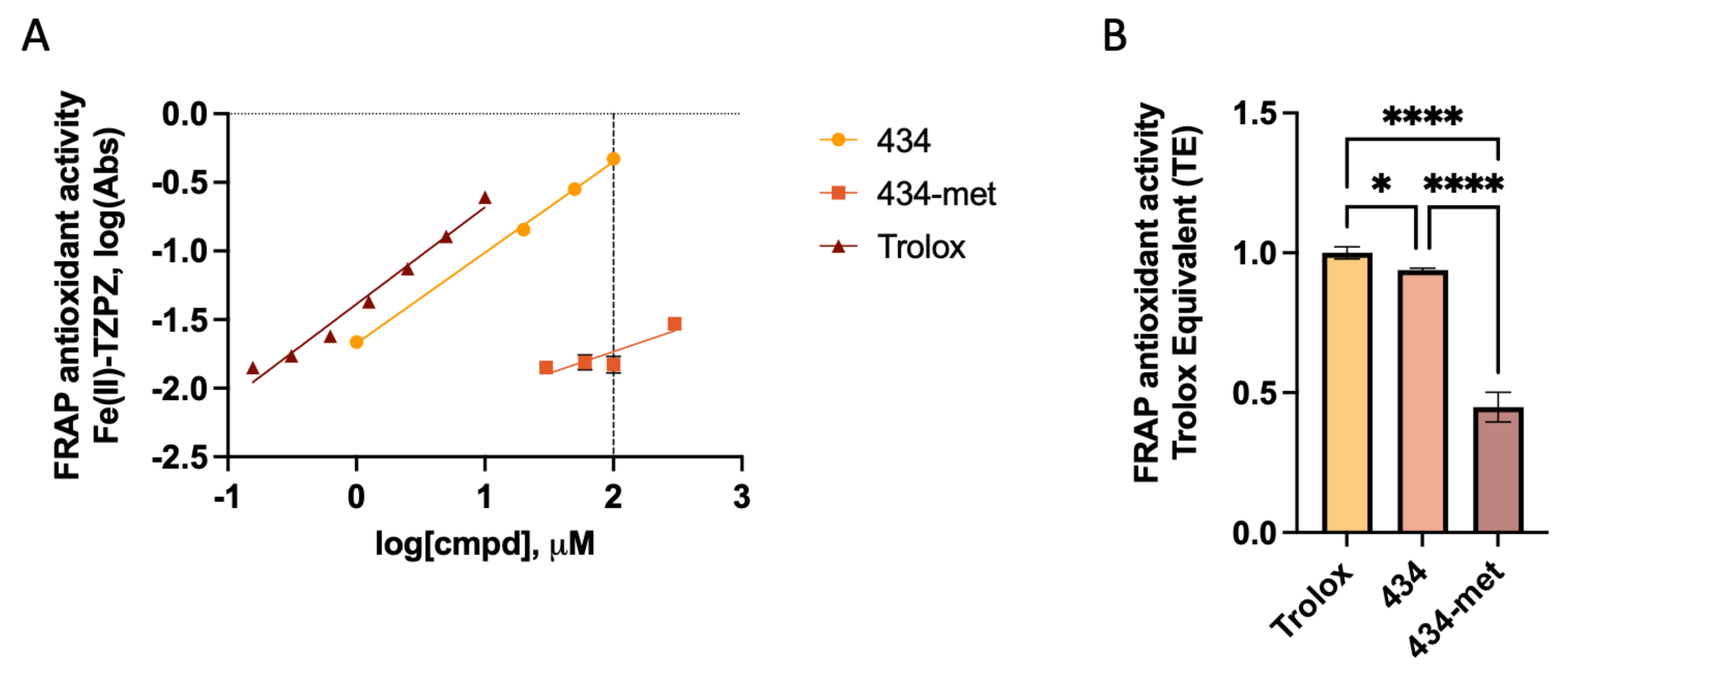
A
FRAP antioxidant activity Fe(II)-TZPZ, log(Abs)
log[cmpd], µM
434
434-met
Trolox
B
FRAP antioxidant activity Trolox Equivalent (TE)

434 has electron transfer capacity determined by FRAP assay. A*, FRAP assay was used to determine the ET capacity of 434 and 434-met in comparison to Trolox. Data are presented as the log transformed absorbance of the reduced Fe(II)-TZPZ complex vs the compound concentration, then fit using linear regression on the log transformed data.* ***B****, The slopes of the linear regression in panel A are plotted for each compound as the Trolox equivalent (TE) (m_{cmpd} / m_{Trolox}) ± SEM of the slopes.*

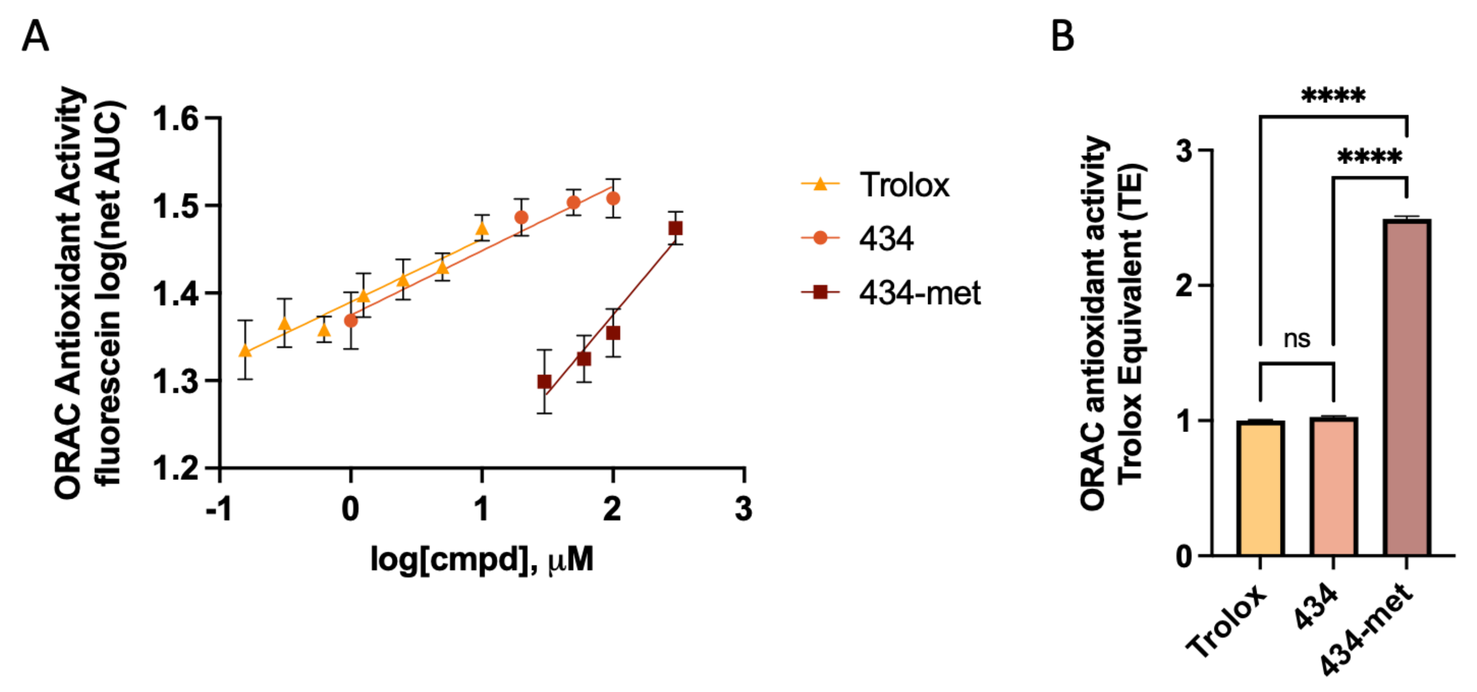
A
ORAC Antioxidant Activity fluorescein log(net AUC)
log[cmpd], µM
Trolox
434
434-met
B
ORAC antioxidant activity Trolox Equivalent (TE)

434 has hydrogen atom transfer capacity determined by ORAC assay. A*, ORAC assay was used to determine the HAT capacity of 434 and 434-met in comparison to Trolox. Data are presented as the log transformed net AUC of fluorescein vs the compound concentration, then fit using linear regression on the log transformed data.* ***B****, The slopes of the linear regression in panel A are plotted for each compound as the Trolox equivalent (TE) (m_{cmpd} / m_{Trolox}) ± SEM of the slopes.*

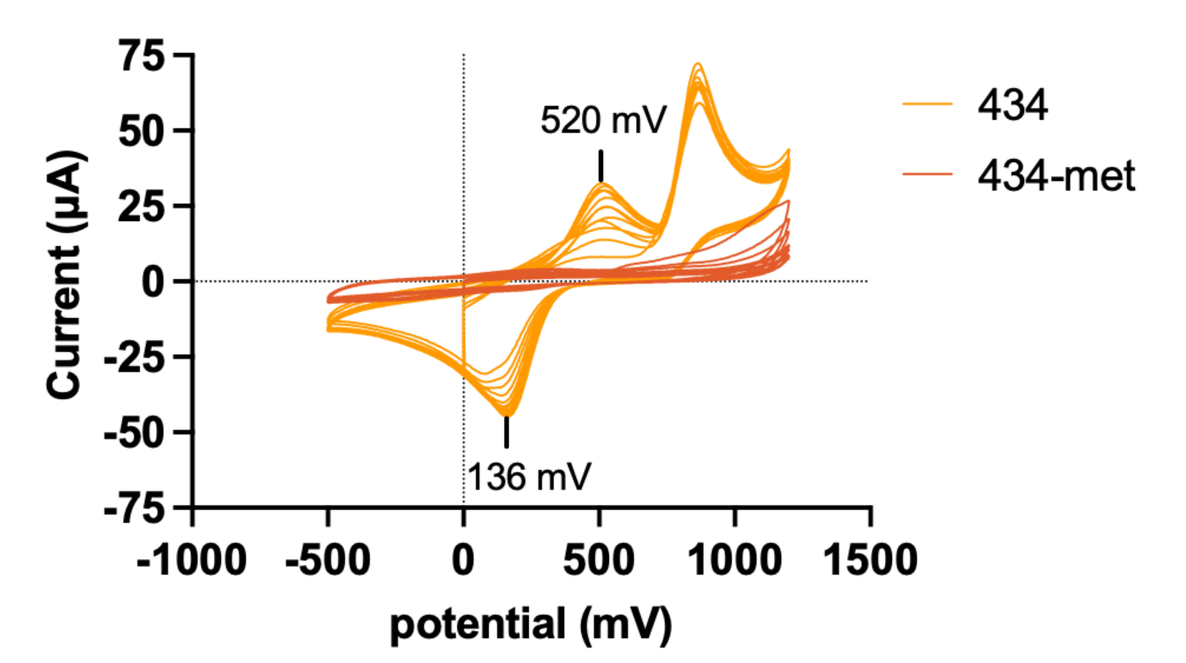
Current (µA)
potential (mV)
520 mV
136 mV
434
434-met

434 has electron transfer capacity in cyclic voltammetry (CV). *5mM solutions of 434 or 434-met in 0.1M KCl, 10% DMSO were analyzed by cyclic voltammetry against a Ag/AgCl electrode. Scans were run at 100mV/s, 6 segments per scan.*

	434 reduction potential (mV)
Mean	348.8
SD	37.16
SEM	18.58
n	4

Reduction potential of ATH434 determined by CV. *Data are the average of 4 independent scans, with 3 replicates each.*

Conclusion

Together, these results suggest that antioxidant activity may be an important contributor to the efficacy of ATH434 in neurodegenerative disorders characterized by excess labile central iron, thus enhancing the efficacy of its moderate iron binding. We conclude that:

- **434 vs Dfp:** 434 possesses potent antioxidant activity that Dfp does *not.* This antioxidant activity supports and protects the mitochondria as shown using TMRM. Dfp *did* however exhibit peroxide scavenging activity, an effect that could provide ROS protection but does not correlate to antioxidant or mitochondrial protective effects as shown with ABTS and TMRM assays. Future studies will focus on comparing mitochondrial activity in cells treated with 434 or Dfp using Seahorse analysis, as well as analyzing the effects on the lipid peroxidation downstream of ROS.
- **434 vs 434-met:** 434 is a more potent antioxidant than 434-met, likely due to its electron transfer capacity. 434-met does *not* have activity in CV or FRAP, assays that specifically target ET, but does have antioxidant activity in ABTS and ORAC which assess HAT activity. Future studies will assess this electron transfer capacity in a cellular context, namely Seahorse analysis of mitochondrial respiration and potential antioxidant effects on lipid peroxidation.

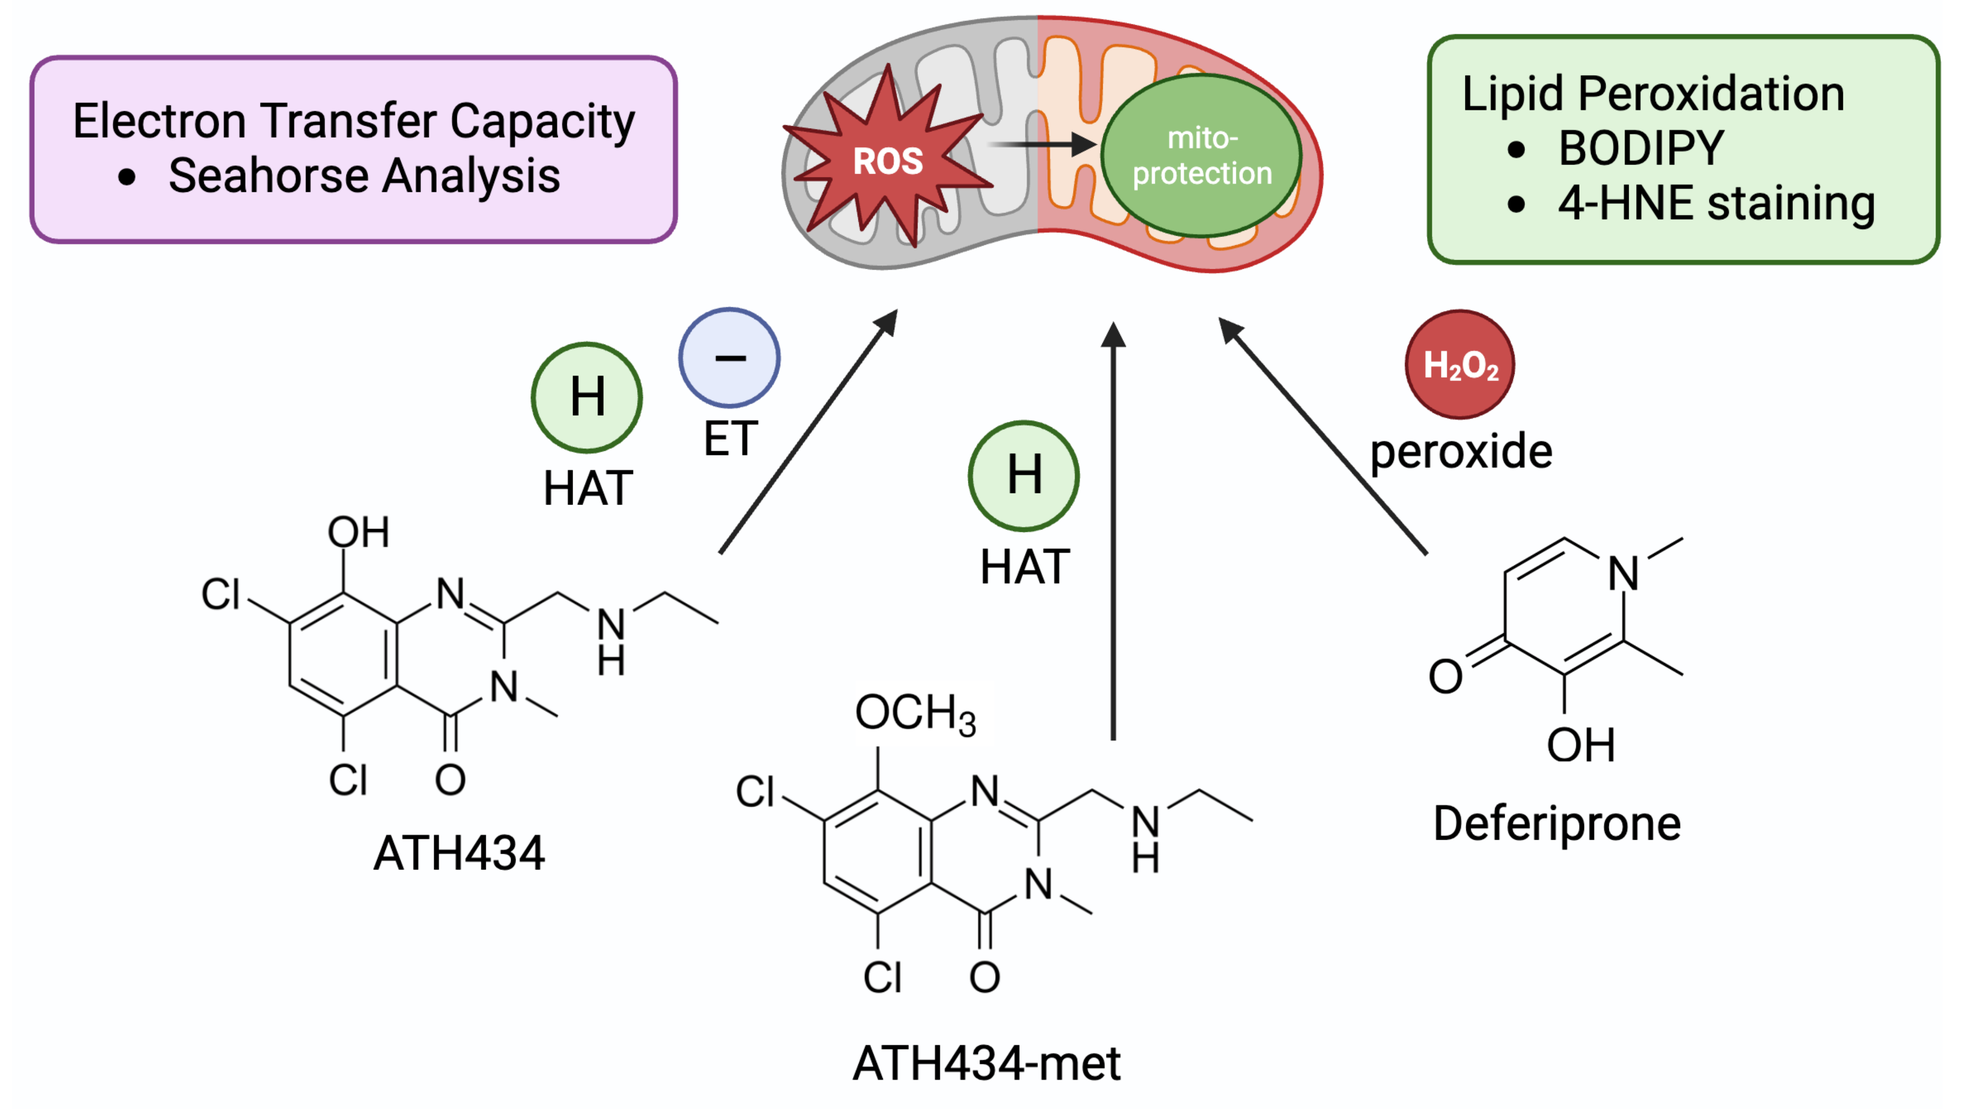
Electron Transfer Capacity
Seahorse Analysis
ROS
mito-protection
Lipid Peroxidation
BODIPY
4-HNE staining
HAT
ET
HAT
peroxide
ATH434
ATH434-met
Deferiprone

References

1. Finkelstein, David et. al. "The novel compound PBT434 prevents iron mediated neurodegeneration and alpha-synuclein toxicity in multiple models of Parkinson's disease." *Acta neuropathologica communications* vol. 5,1 (2017):53. doi:10.1186/s40478-017-0456-2

2. Zhang, Ming-Xia et al. "Design, synthesis, and antimicrobial evaluation of hexadentate hydroxypyridinones with high iron(III) affinity." *Chemical Biology & Drug Design* vol. 84,6 (2014): 659-68. doi:10.1111/cbdd.12358

3. Ma, Yongmin et al. "Synthesis and characterizations of pyridazine-based iron chelators." *Dalton Transactions* (Cambridge, England : 2003) vol. 43,45 (2014): 17120-8. doi:10.1039/c4dt02687j

4. Devos, David et al. "Trial of Deferiprone in Parkinson's Disease." *The New England Journal of Medicine* vol. 387, 22 (2022): 2045-2055. doi:10.1056/NEJMoa2209254

University at Buffalo The State University of New York

Department of Biochemistry
Jacobs School of Medicine and Biomedical Sciences
buffalo.edu